1173. Prevalence of pterygium in the United States: a claims-based analysis

Abu Abraham[1], Gregory Brooks[1], Christopher Dadas[1], John Hovanesian[2], Jessica Lee[3], Jinsong Ni[1], Scott Whitcup[2]

[1]Cloudbreak Pharmaceutical, Irvine, CA, US; [2]UCLA Stein Eye Institute, David Geffen School of Medicine at UCLA, Los Angeles, CA, US; [3]Syneos Health Consulting, South San Francisco, CA, US

Purpose

- Pterygium is a common fibrovascular growth extending from the conjunctiva to the corneal surface that ranges in severity from mild to moderate eye irritation and redness to severe presentations (**Figure 1**)[1,2]
- As it advances, pterygium can compromise vision if it causes irregular astigmatism or corneal surface disease or if the lesion grows over the pupil[1]
- Available topical treatments include corticosteroids and other anti-inflammatory drugs to manage symptoms, and surgery may be considered for patients with severe cases[1-3]
- Age and exposure to ultraviolet (UV) radiation are known risk factors for the development of pterygium[4,5]
- The prevalence of pterygium in the United States (US) is estimated at 15 million people, but the condition likely is underdiagnosed[4,6]
- Here we present a claims analysis to characterize the patient population diagnosed with pterygium in the US

Figure 1. Image of a typical nasal fibrovascular pterygium.

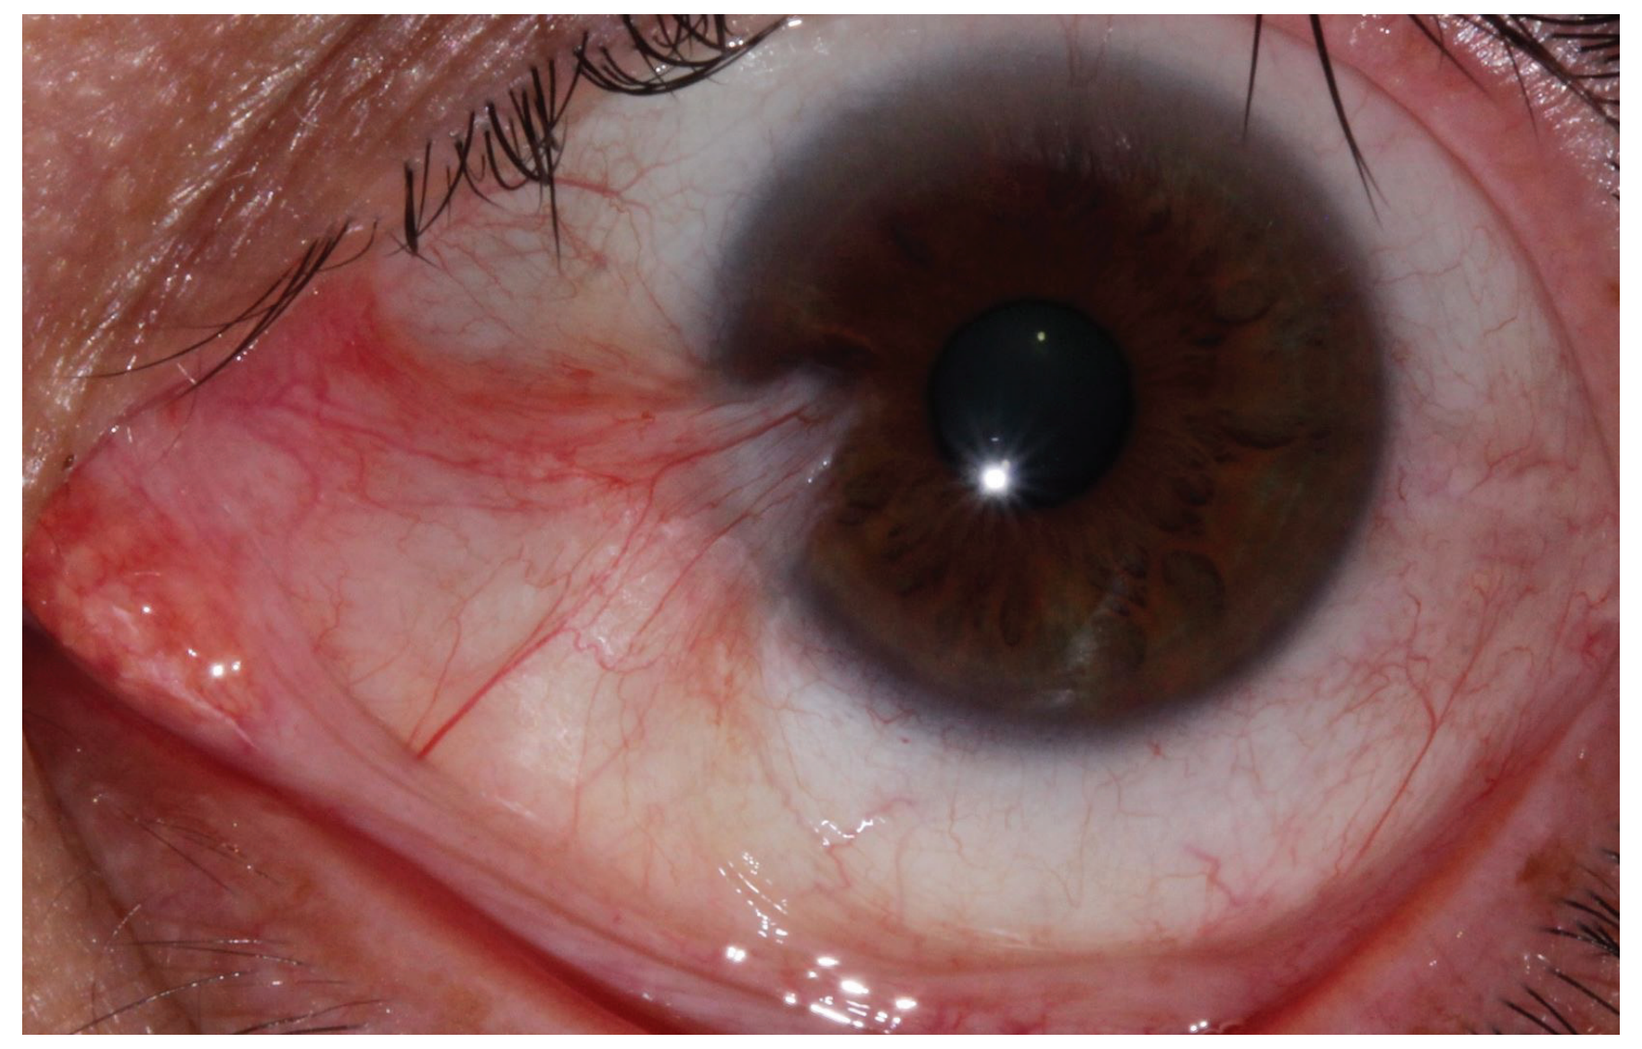

Image courtesy of Abu Abraham, MD.

Methods

- Medical claims data were collected from 2016 through 2021 from the Komodo database to determine pterygium prevalence and from 2018 and 2019 from the Compile database to determine the principal care providers for the condition
- De-identified electronic medical record (EMR) data from 2016 through 2021 were collected from the TriNetX Research Network database to analyze treatment regimens
 - The data used in this study were collected on 2/23/22 from the TriNetX USA Network, which provided access to EMR (diagnoses, procedures, medications, laboratory values, genomic information) from ~142 million patients from 77 healthcare organizations
 - TriNetX is compliant with the Health Insurance Portability and Accountability Act, the US federal law that protects the privacy and security of heathcare data, and any additional privacy regulations applicable to the contributing healthcare organization
- Because this study used only de-identified patient records and did not involve the collection, use, or transmittal of individually identifiable data, this study was exempted from Institutional Review Board approval
- For all analyses, pterygium was defined as *International Classification of Diseases, Tenth Revision (ICD-10)* code 11.00, including all variations
 - Search terms were H11.00, H11.01, H11.02, H11.03, H11.04, H11.05, H11.06, H11.031, H11.032, H11.033, H11.039, H11.061, H11.062, H11.063, H11.069, H11.051, H11.052, H11.053, H11.059, H11.001, H11.002, H11.003, H11.009, H11.011, H11.012, H11.013, H11.019, H11.041, H11.042, H11.043, H11.049, H11.021, H11.022, H11.023, H11.029
- We calculated the period prevalence as the number of patients with pterygium in the Komodo database between 2016 and 2021 divided by the average population during this period in each of the 50 states, the District of Columbia, and Puerto Rico
 - The number of patients with pterygium based on claims in the Komodo database between 2016 and 2021 was derived for each state, the District of Columbia, and Puerto Rico
 - The average population for each state, the District of Columbia, and Puerto Rico was calculated from the total population estimated by the American Community Survey in 2016, 2017, 2018, 2019, and 2021 and the total population from the 2020 US census
 - Results are presented using descriptive statistics. Data were analyzed using Microsoft Excel

Results

- The claims analysis indicates that there are >2 million patients diagnosed with pterygium in the US (**Figure 2**)

Figure 2. Methodology used to calculate the diagnosed prevalence of pterygium.

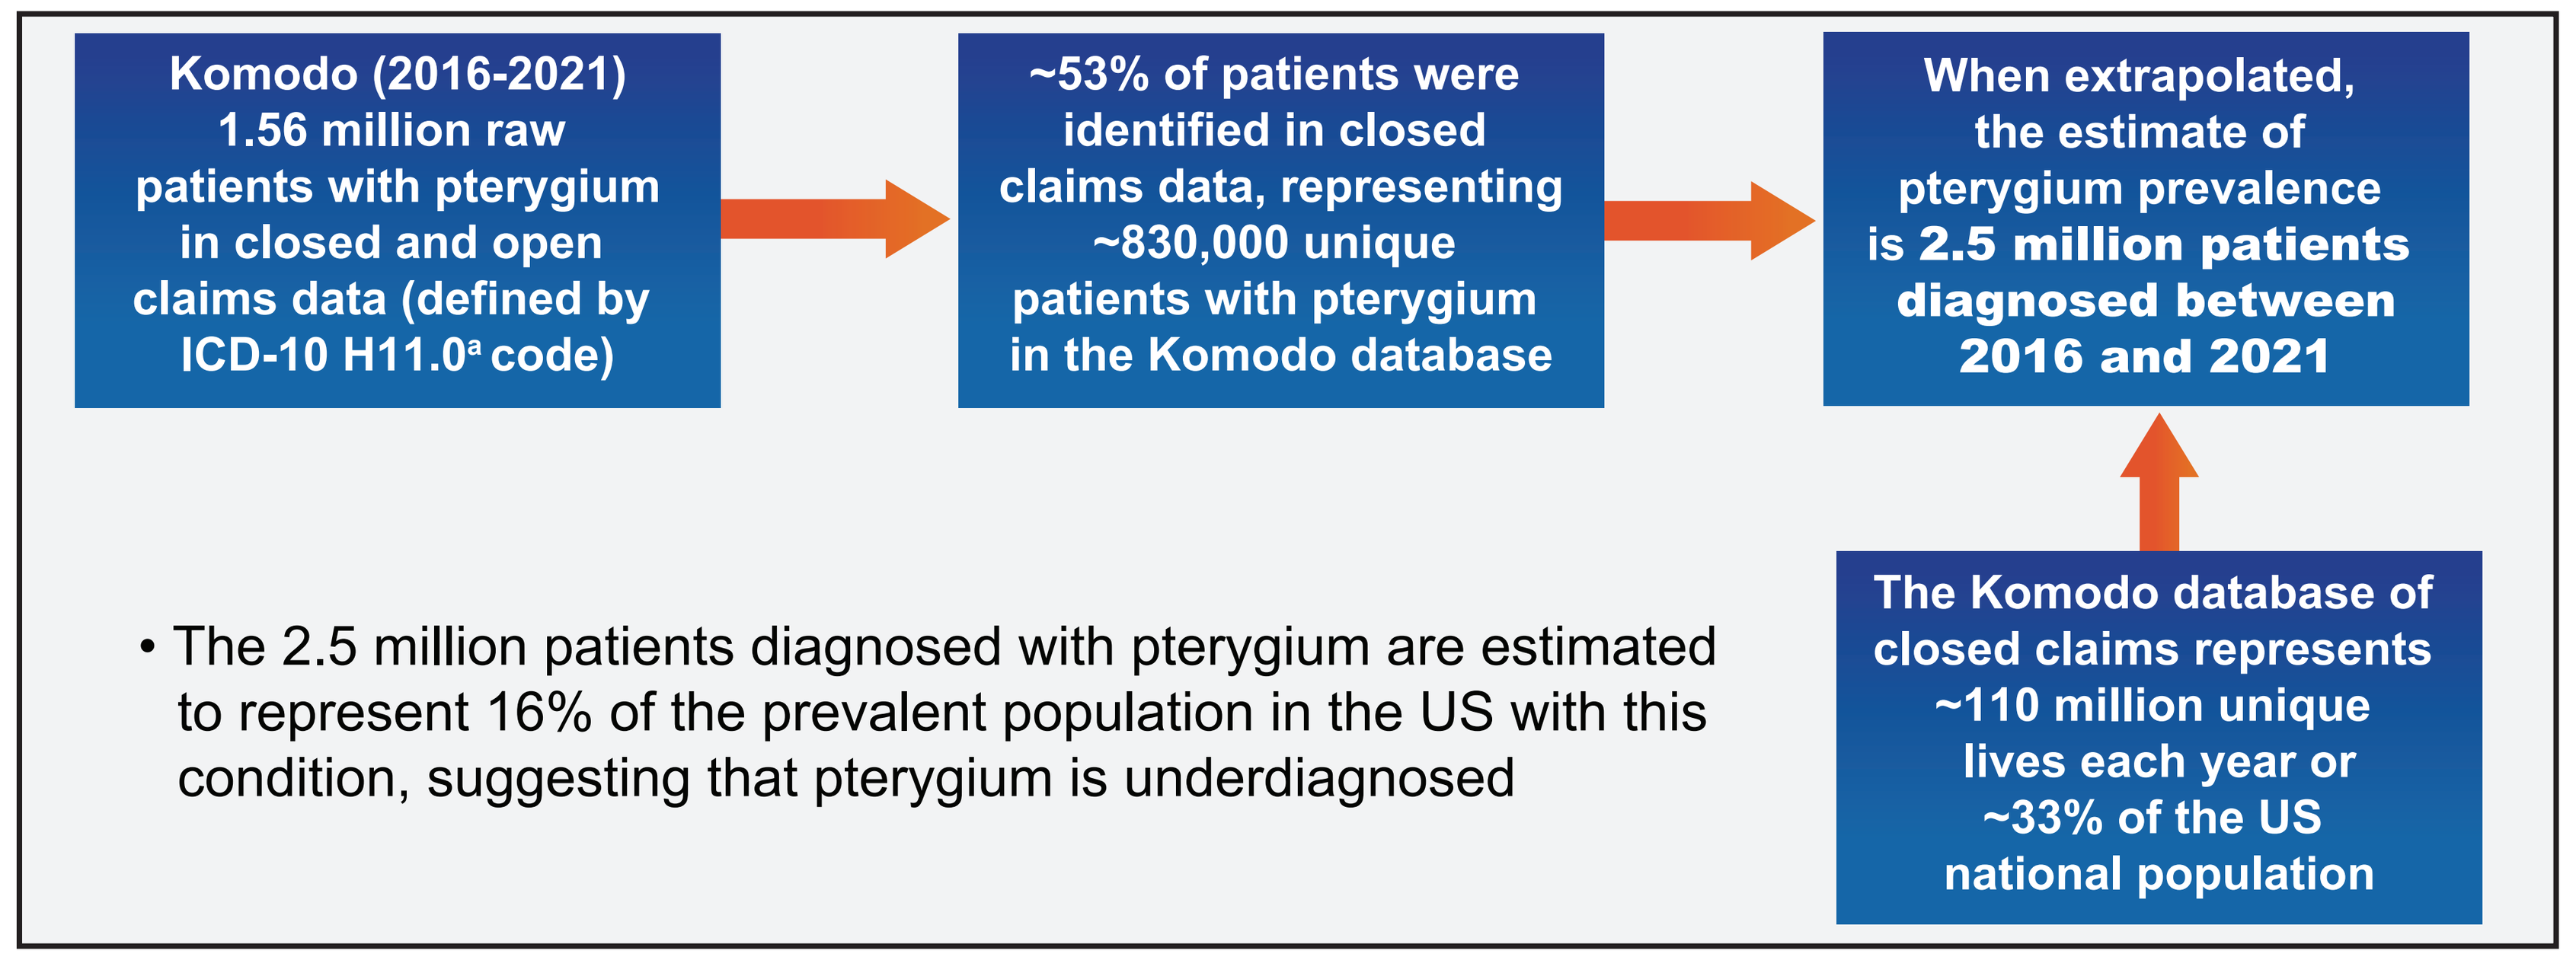
Komodo (2016-2021) 1.56 million raw patients with pterygium in closed and open claims data (defined by ICD-10 H11.0[a] code)
~53% of patients were identified in closed claims data, representing ~830,000 unique patients with pterygium in the Komodo database
When extrapolated, the estimate of pterygium prevalence is 2.5 million patients diagnosed between 2016 and 2021
The Komodo database of closed claims represents ~110 million unique lives each year or ~33% of the US national population
• The 2.5 million patients diagnosed with pterygium are estimated to represent 16% of the prevalent population in the US with this condition, suggesting that pterygium is underdiagnosed

[a]H11.0 included all variations of the *ICD-10* code for pterygium.
ICD-10, International Classification of Diseases, Tenth Revision.
© 2023 Komodo Health, Inc. All rights reserved. Reprinted with permission.

- Pterygium is primarily managed by ophthalmologists and optometrists (**Figure 3**)

Figure 3. Pterygium patient volume distribution by specialty, June-December 2019.

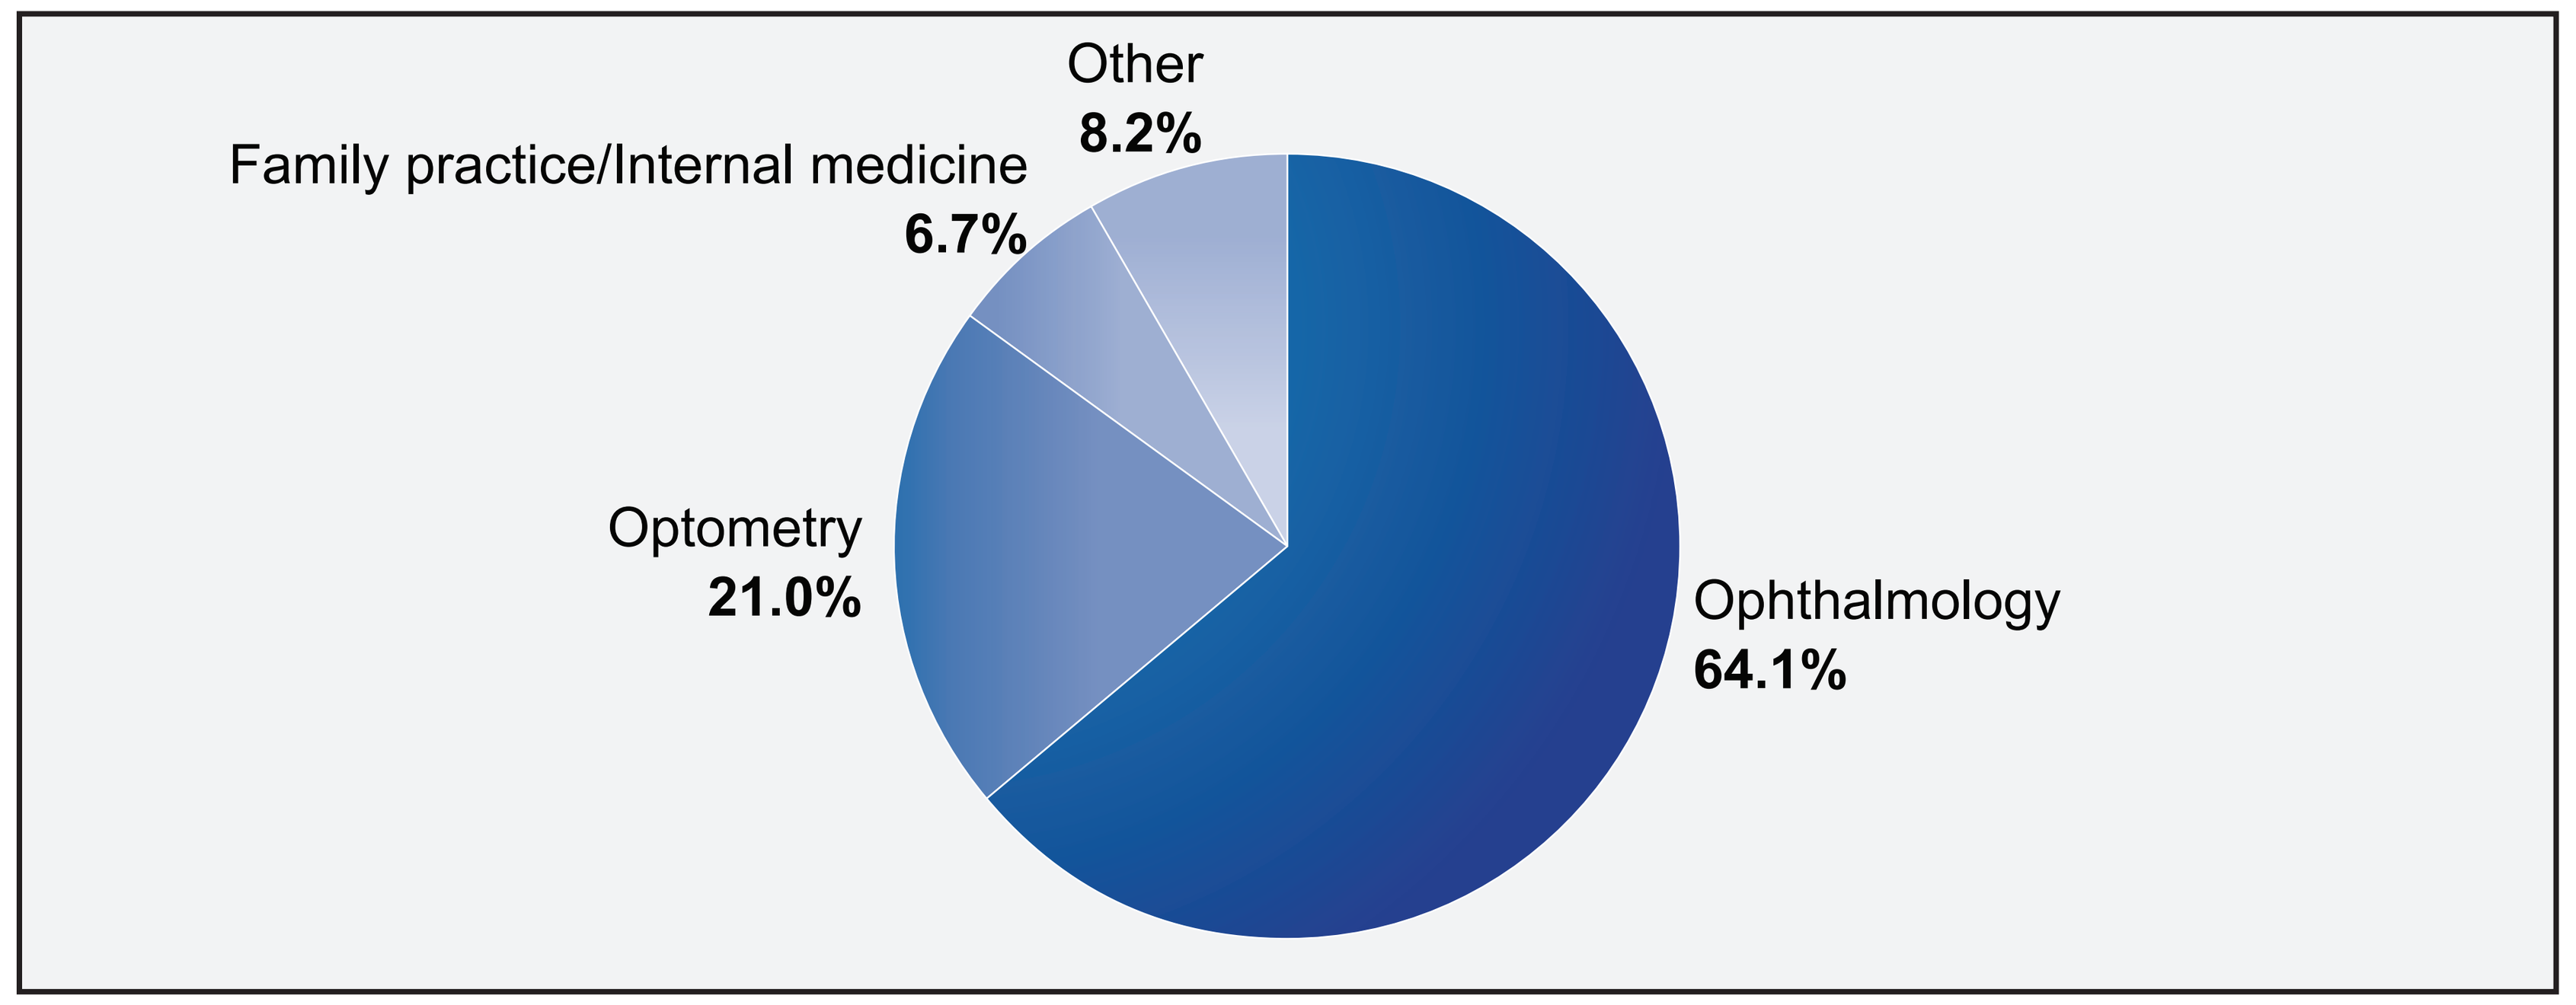
Other
8.2%
Family practice/Internal medicine
6.7%
Optometry
21.0%
Ophthalmology
64.1%

 - Collectively, eye care professionals care for ~85% of patients with pterygium
 - Although there are more optometrists in the US,[7,8] ophthalmologists were the most common care providers for patients with pterygium
- The majority of patients are prescribed topical treatments, including corticosteroids and nonsteroidal anti-inflammatory drugs (**Table**)

Table. Medical management of pterygium

Therapy group	Treated patients with pterygium (%)
NSAIDs	35
Corticosteroid	47
Cyclosporine or lifitegrast	3
Surgery	13
Any treatment	59

NSAID, nonsteroidal anti-inflammatory drug.

 - Lubricants (e.g., artificial tears) are also commonly used by patients with pterygium,[2] but these over-the-counter treatments were not captured in patient EMRs

- The period prevalence (2016-2021) of patients with pterygium in the Komodo database for each state highlights areas with greater UV exposure and population centers (**Figure 4**)
 - The states with the highest period prevalence (2016-2021) of patients with pterygium in the Komodo database per 1000 people in the state population were Hawaii (15.3 per 1000), California (9.8 per 1000), New Mexico (9.5 per 1000), New York (8.7 per 1000), and Florida (7.9 per 1000). Puerto Rico had 7.0 patients with pterygium in the Komodo database per 1000 people
 - 37 states had fewer than 4 patients with pterygium in the Komodo database per 1000 people in the state population

Figure 4. Period prevalence (2016-2021) of patients with pterygium in the Komodo database per 1000 people in the state population.

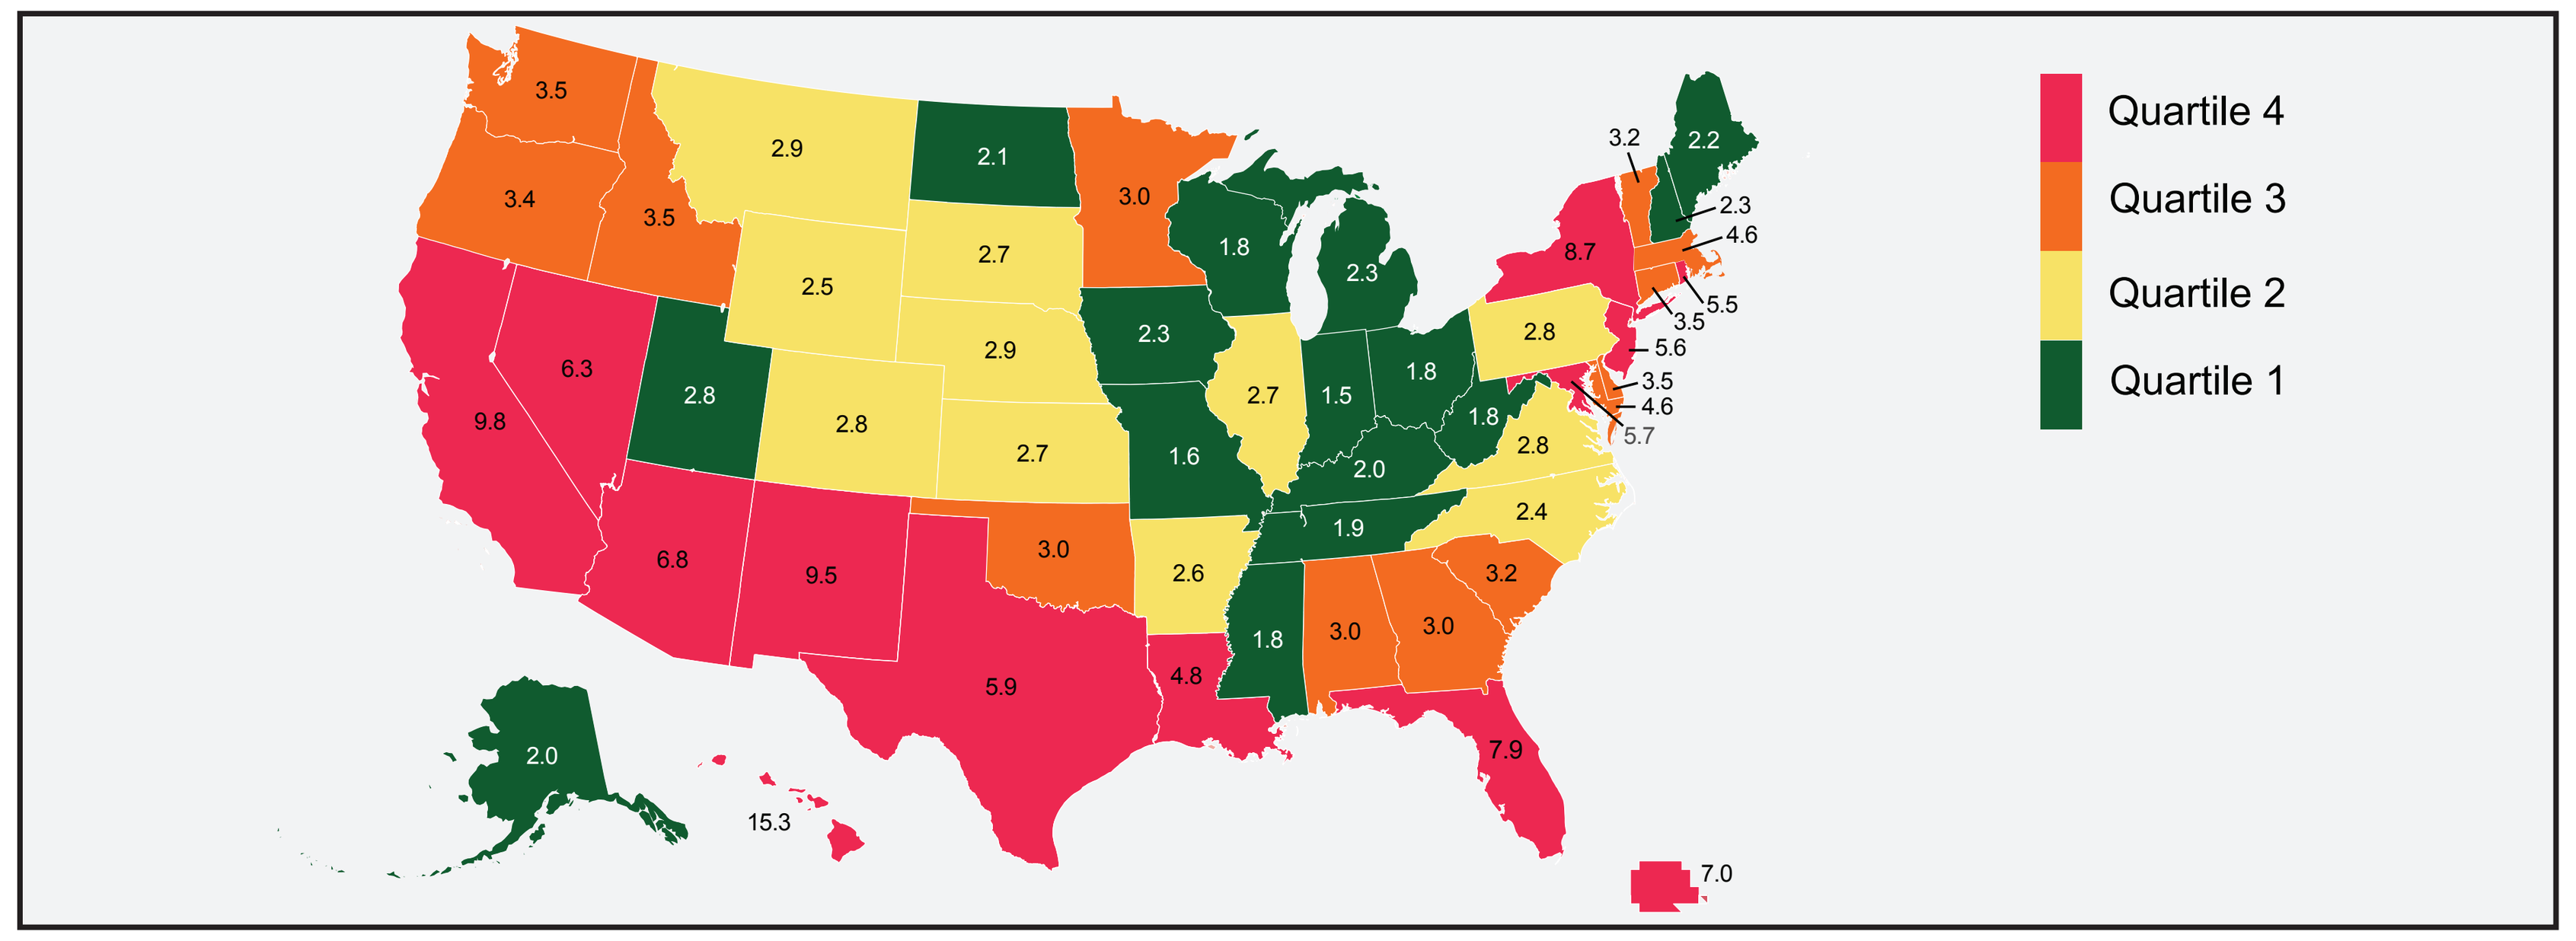
Quartile 4
Quartile 3
Quartile 2
Quartile 1
3.5
2.9
2.1
3.0
3.2
2.2
3.4
3.5
2.7
1.8
2.3
8.7
2.3
4.6
2.5
2.3
5.5
3.5
2.8
5.6
6.3
2.9
3.5
4.6
2.8
2.8
2.7
1.5
1.8
9.8
1.8
5.7
2.7
1.6
2.0
2.8
2.4
1.9
3.0
6.8
9.5
2.6
3.2
1.8
3.0
3.0
5.9
4.8
2.0
7.9
15.3
7.0

© 2023 Komodo Health, Inc. All rights reserved. Reprinted with permission.

Conclusions

- In an analysis of closed insurance claims in the US, approximately 2.5 million individuals with pterygium sought care between 2016 and 2021
- Because of the high prevalence rate, it is believed that this condition is underdiagnosed among eye care professionals
 - The contrast between the estimate of 2.5 million diagnosed cases among a projected prevalence of 15 million people in the US suggests that most cases are not included in insurance claims, potentially because of a lack of available treatments
 - There is an unknown, but significant, population of immigrants to the US from high–pterygium-risk countries in Latin America who often have high-risk occupations (e.g., farmworkers, construction workers, other outdoor workers) for pterygium development[4,9]
 - This population may not be accessible in a claims-based analysis but may increase the actual prevalence significantly
- Eye care professionals (ophthalmologists and optometrists) are the principal healthcare providers for pterygium
- Topical agents (eyedrops) are the most common treatments for pterygium, but they do not address the underlying cause of the disease
- For advanced pterygium affecting vision, surgery is a treatment option, but this approach is not preferred by patients because of the perception of morbidity and pain and the chance for recurrence[2,10]
- There is an unmet medical need for the development of an effective topical therapy in addition to understanding the rate of diagnosis and treatment for this condition[11]

Acknowledgments

This study was supported by Cloudbreak Pharmaceutical. Medical writing assistance was provided under the direction of the authors by Katherine Stevens-Favorite, PhD, of Cadent, a Syneos Health group company, and was supported by Cloudbreak Pharmaceutical.

Disclosures

Abu Abraham is an employee of Cloudbreak Pharmaceutical. Jinsong Ni is the founder and a current employee of Cloudbreak Pharmaceutical and holds relevant patents. Gregory Brooks, Christopher Dadas, John Hovanesian, Jessica Lee, and Scott Whitcup are consultants for Cloudbreak Pharmaceutical.

References

1. Shahraki T, et al. *Ther Adv Ophthalmol*. 2021;13:25158414211020152. **2.** Chu WK, et al. *Eye*. 2020;34(6):1047-1050. **3.** Frucht-Pery J, et al. *Am J Ophthalmol*. 1999;127:148-152. **4.** Modenese A, et al. *Int J Environ Res Public Health*. 2018;15:2063. **5.** Liu L, et al. *BMJ Open*. 2013;3:e003787. **6.** Lucas R, et al. *Environmental Burden of Disease Series, No. 13: Solar Ultraviolet Radiation: Global Burden of Disease from Solar Radiation*. World Health Organization; 2006. Accessed February 27, 2023. https://www.who.int/publications/i/item/9241594403. **7.** Dang S, et al. *Invest Ophthalmol Vis Sci*. 2021;62(8):1724. **8.** US Bureau of Labor Statistics. Occupational Outlook Handbook. *Optometrists*. Updated September 8, 2022. Accessed February 27, 2023. https://www.bls.gov/ooh/healthcare/optometrists.htm. **9.** West S, Muñoz B. *Br J Ophthalmol*. 2009;93:1287-1290. **10.** Gotkas S, et al. *Clin Exp Optom*. 2017;100:595-597. **11.** Wanzeler ACV, et al. *Clin Ophthalmol*. 2018;12:833-837.

Poster presented at the ARVO Annual Meeting; April 23-27, 2023; New Orleans, LA